VALIDATION OF A PATIENT REPORTED EXPERIENCE MEASURE IN PATIENTS WITH RHEUMATOID ARTHRITIS

CQ RA Commissioning for Quality in Rheumatoid Arthritis

Roche

Marwan Bukhari,[1] Peter Jones,[2] Ifeanyi Sargeant,[3] Maureen Cox,[4] Alison Elliott,[5] Suzanne Bullock,[6] Anne O'Brien[2] and Ailsa Bosworth,[7]

[1]Royal Lancaster Infirmary, [2]Keele University, [3]IS medical communications, [4]Nuffield Orthopaedic centre, [5]Roche Pharmaceuticals,[6]Lancaster University, [7]National Rheumatoid Arthritis Society

Background/Purpose

- Improving patient experience is important in all diseases, but most important in patients with chronic rheumatic diseases like rheumatoid arthritis
- To date there have been no specific patient reported experience measures (PREMs) in rheumatology. This would be a method of ascertaining that the patient has an optimum experience when attending for their care
- A multi-disciplinary group, commissioning for quality in rheumatoid arthritis (CQRA) was established to develop the first PREM in RA
- This group was supported under the terms of a joint working agreement with Roche Products Ltd. (*Ailsa Bosworth, Chief Executive, NRAS, Joint Chair Rheumatology Futures Group; Graeme Johnston, NRAS, Chair of the Trustees; Dr. Marwan Bukhari, Consultant Rheumatologist, Royal Lancaster Infirmary; Maureen Cox, Clinical Nurse Specialist in Rheumatology, Nuffield Orthopaedic Centre (NHS Trust); Anne O'Brien, Lecturer in Physiotherapy - Keele University Staffordshire; Prof. Peter Jones, Senior Statistician at the Health Services Research Unit, and Emeritus Professor of Statistics, Keele University; Paul Carroll, Assistant Director Commissioning, Ashton, Leigh and Wigan PCT; Dr. Alan Nye, Director Pennine MSK Partnership; Alison Elliott, Strategic Funding Manager-Speciality Care, Roche Products Limited; Ify Sargeant, Medical Writer, ismedica Ltd.*)
- A series of focus groups were held with patients from the UK National Rheumatoid Arthritis Society (NRAS) to determine which elements of the patient experience was deemed most important
- This was mapped against the UK Department of Health Patient Experience Framework which comprises 8 domains
- A questionnaire was then developed using the same 8 domains, but with questions developed specifically relating to RA and Rheumatology Services
- This questionnaire was then piloted across ten UK sites
- The final question asked respondents to evaluate their overall level of care
- All questions were graded from strongly agree to strongly disagree

Figure 1. Map of UK showing participating unit locations (white dots)

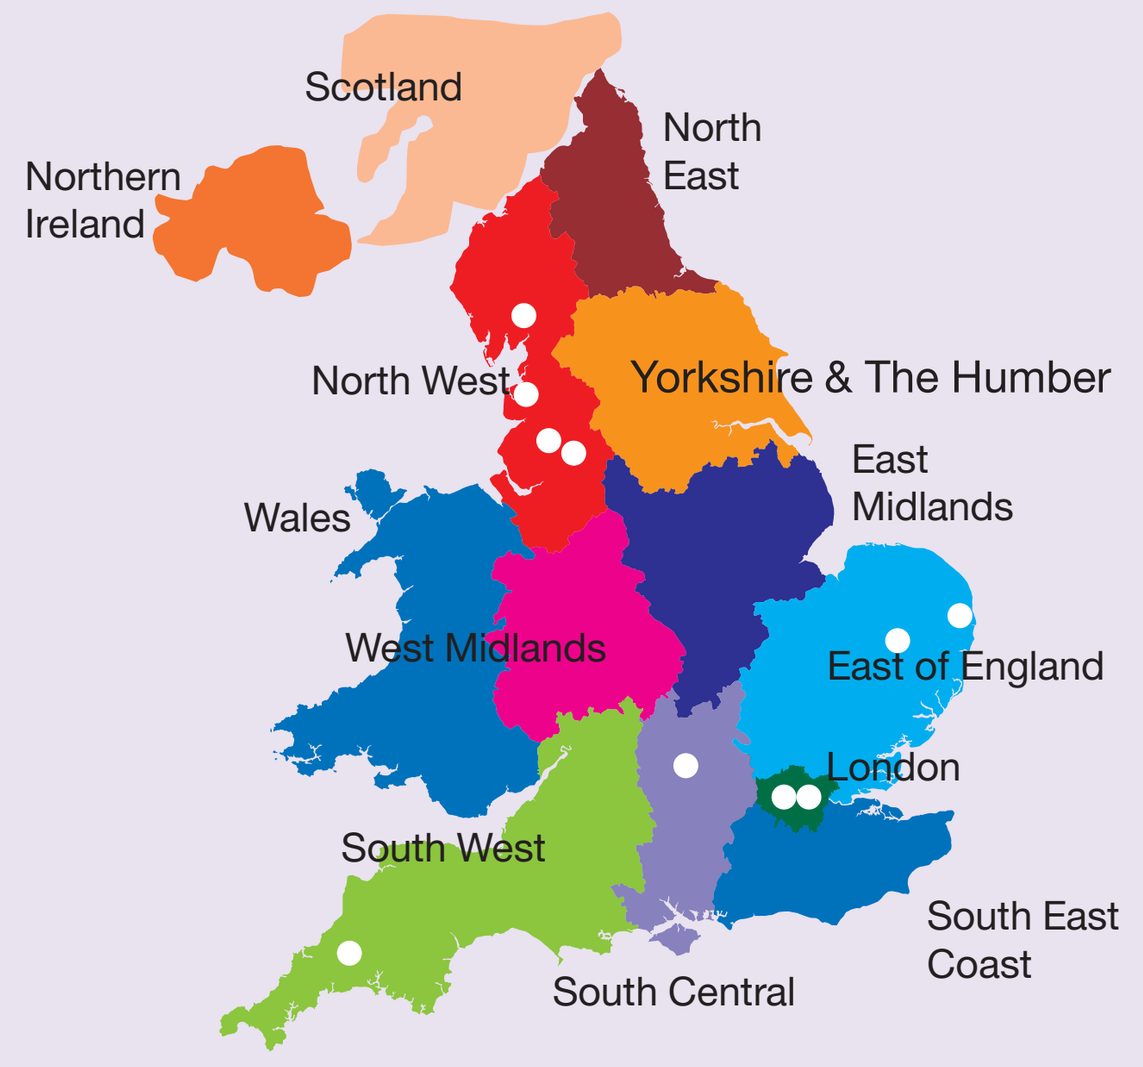
Scotland
Northern Ireland
North East
North West
Yorkshire & The Humber
East Midlands
Wales
West Midlands
East of England
London
South West
South East Coast
South Central

Aim

To determine the construct validity of the PREM questionnaire developed by the CQRA group, to determine it's ability to measure the patient experience

Methods

- Cronbach's alpha[1] (Bland & Altman (1997) was chosen to measure construct validity. It is commonly used over ordinal scales to measure internal consistency within a domain or how closely the individual questions are related. A value of at least 0.7 is regarded as satisfactory. The percentage agreement with overall care over the 5 point scale for each questions within a domain was also calculated.
- Additionally, for each question the percentage agreement with the overall assessment on the five point scale was calculated
- In case of multiple questions per domain, the responses are shown as a range

Results

- 524 patients were included in the analysis
- Median age was 65 years (IQR 55,80 years)
- 377 (72%) were female
- Median RA disease duration was 8 years (IQR 3.5, 15 years)
- The Cronbach alpha co-efficients within the multi-question domains and their percentage agreement with the question on overall care are shown in table1
- This demonstrated good within domain agreement and agreement of each domain with overall experience of care

Table 1. Result of Cronbach's alpha analysis and their agreement with overall care

Domain	Number of questions	Alpha within domain	Agreement with overall care (%)
Needs and preferences	5	0.90	64.5–67.2
Co-ordination of care	4	0.87	59.1–69.1
Information about care	4	0.75	22.5–66.4
Daily living	2	0.61	33.2–53.4
Emotional support	2	0.84	53.5–68.9
Family and friends	1	–	61.8
Access to care	1	–	70.4

Conclusion

- The PREM has good construct validity and is a valid tool for measuring RA patient experience
- Some domains have higher agreement with overall patient experience
- This could provide a useful future tool for measuring patient experience
- Modification of the tool to use in other rheumatic conditions is underway

Reference

1, M. Bland & D. Altman Statistics notes: Cronbach's alpha BMJ 1997;314:572

Acknowledgements:

With thanks to units/hospitals participating in the survey who returned questionnaires for pooled analysis: Pennine MSK Partnership Ltd, Oldham; West Suffolk Hospital, Bury St Edmunds; James Paget University Hospitals NHS Trust, Great Yarmouth; Royal Cornwall Hospital, Truro; Guys and St Thomas' Hospital, London; Nuffield Orthopaedic Centre, Oxford; Salford Royal NHS Foundation Trust, Salford; Whipps Cross University Hospital, London; Queen Victoria Hospital, Blackpool; Royal Lancaster Infirmary, Lancaster.

Roche products Ltd are supporting this project by providing project management, facilitation, medical writing support and printing costs.

The PREM questionnaire

Section	Statement	Strongly agree	Agree	Neither agree nor disagree	Disagree	Strongly disagree
1. Your needs and preferences	a) Whenever I attended a clinic, I felt that I was treated respectfully as an individual	○	○	○	○	○
	b) I was involved as much as I wanted to be in decisions about my treatment and care	○	○	○	○	○
	c) My personal circumstances (see note 1 below) and preferences were taken into account when planning and deciding on my treatment and care	○	○	○	○	○
	d) I was given information in a way that I could understand	○	○	○	○	○
	e) I was given enough information to help me make decisions about my treatment	○	○	○	○	○
2.Co-ordination of care and communication Care across departments	a) I was made aware that there is a team of health professionals (see note 2 below) looking after me	○	○	○	○	○
	b) When I needed help I was able to access different members of my health team	○	○	○	○	○
	c) There is a member of my health team who can help me to see other specialists in the team if I need to	○	○	○	○	○
	d) I feel that the people I see at the clinic are fully up to date with my current situation	○	○	○	○	○
3. Information, education and self-care	a) I feel that I was given information at the time I needed it	○	○	○	○	○
	b) I feel that I have a good understanding of the treatments I am on or being offered	○	○	○	○	○
	c) I have been told about patient organisations or groups that can help me	○	○	○	○	○
	d) I have been offered an opportunity to attend a self-management programme suitable to my needs	○	○	○	○	○
4. Daily living and physical comfort	a) I feel that my rheumatoid arthritis is being controlled enough to let me get on with my daily life and usual activities	○	○	○	○	○
	b) If I have had a 'flare' (when my symptoms get much worse), I have been able to get help quickly	○	○	○	○	○
5. Emotional support	a) I feel able to approach a member of my health team to discuss any worries about my condition and my treatment or their effect on my life	○	○	○	○	○
	b) I feel able to discuss personal or intimate issues about relationships with my health team if I want to	○	○	○	○	○
6. Family and friends	a) I feel able to take members of my family to outpatient appointments to become involved in decisions about my care if I want to	○	○	○	○	○
7. Access to care	a) At appointments, I feel that I have enough time with the healthcare professional to cover everything I want to discuss	○	○	○	○	○
	b) I have had clinic appointments cancelled unexpectedly	○ Yes	○ No			
	c) *If yes, how long have you had to wait for a new appointment?*	○ <1 week	○ 1–3 weeks	○ 4–6 weeks	○ 7–12 weeks	○ >12 weeks
	d) I have needed extra treatment or a change of treatment	○ Yes	○ No			
	e) *If yes, how long did it take for this to happen?*	○ <1 week	○ 1–3 weeks	○ 4–6 weeks	○ 7–12 weeks	○ >12 weeks
8. Overall experience of care	a) Overall in the past year, I have had a good experience of care for my rheumatoid arthritis	○	○	○	○	○

The length of time you have had rheumatoid arthritis, your age, sex and ethnic background may affect the kind of care you have experienced and affect your answers to the questions. To help us fully understand your answers, please fill in the section below.

How long have you had rheumatoid arthritis?

○ Less than 2 years ○ Between 2 and 5 years ○ Between 6 and 10 years ○ More than 10 years

How old are you?

○ Under 30 ○ 30 to 40 ○ 41 to 50 ○ 51 to 60 ○ 61 to 70 ○ Over 70

How old are you?

○ male? ○ female?

What is your ethnic group?

○ White ○ Mixed ○ Asian or Asian British ○ Black, African, Carribean or Black British ○ Other ○ Prefer not to say

Thank you for taking the time to fill in this questionnaire.

October 2013

RXUKHCMR00511a

Marwan's Poster Oct 2013_v1e.indd 1

14/10/2013 18:03